Cellular Hormone Metabolism is Critical for Canonical Androgen Receptor Antagonist Activity

Suriyan Ponnusamy[1], Wendy Effah[1], Thirumagal Thiyagarajan[1], Dong-Jin Hwang[2], Yali He[2], James B. Breitmeyer[3], Gunnar F. Kaufmann[3], Duane D. Miller[2,4], Ramesh Narayanan[1,4]

[1] Department of Medicine, [2] Department of Pharmaceutical Sciences, University of Tennessee Health Science Center, Memphis, TN., [3] Oncternal Therapeutics Inc. San Diego, CA, [4] UTHSC Cancer Center

THE UNIVERSITY OF TENNESSEE HEALTH SCIENCE CENTER

Introduction

- Androgen receptor (AR) is the primary therapeutic target expressed in over 85% of prostate cancer (PC), a leading cause of cancer-related deaths in the US.
- PC that relapses from hormonal treatment (castration-resistant prostate cancer (CRPC)) is aggressive and contributes to the majority of PC-related deaths.
- Second-generation AR antagonists and androgen-synthesizing enzyme inhibitors have significantly extended survival of CRPC patients.
- After a brief period of response, resistance to AR-pathway inhibitors develops frequently via various mechanisms.
- **The objective of this work is to discover the mechanisms by which resistance to AR antagonists develops and if N-terminus domain (NTD)-binding AR inhibitors can bypass the resistance mechanism.**

Summary

- Canonical AR antagonists (1st and 2nd generation) upregulate androgen metabolizing enzymes UGT2B15, 17, and 28 to reduce intracellular androgen levels.
- Knock-down of UGT2B expression impairs AR antagonist activity in vitro.
- Decreased UGT2B expression in vivo leads to reduced canonical AR antagonist efficacy.
- Dual Action AR Inhibitors (DAARIs) UT-34 (ONCT-534) and UT-105 (ONCT-505) that bind to AR NTD inducing AR protein degradation, act independently of UGT2B expression levels.

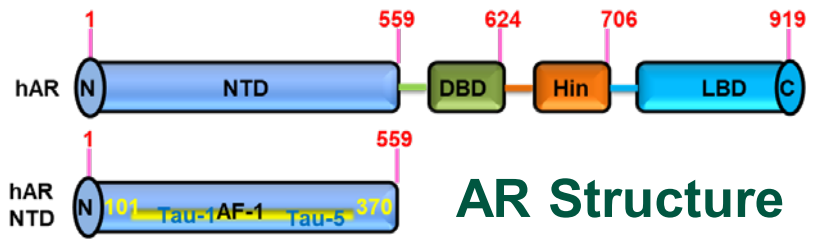
1
559
624
706
919
hAR
N
NTD
DBD
Hin
LBD
C
1
559
hAR NTD
101
Tau-1
AF-1
Tau-5
370

AR Structure

DAARIs Bind to AR NTD and Degrade AR Protein

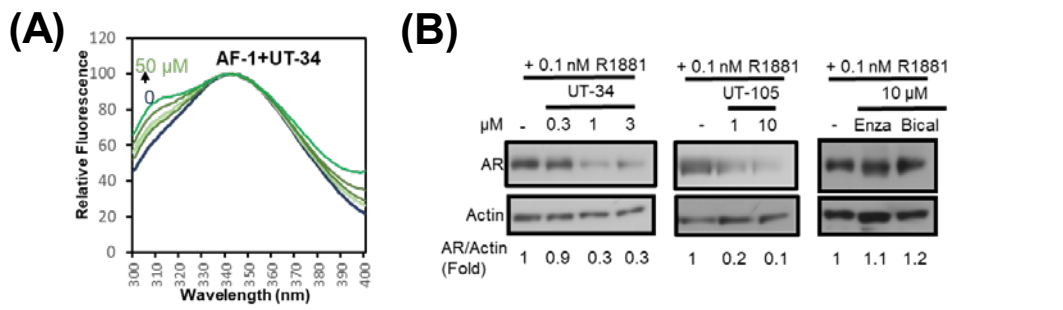
(A)
AF-1+UT-34
50 μM
0
Relative Fluorescence
Wavelength (nm)
(B)
+ 0.1 nM R1881
UT-34
+ 0.1 nM R1881
UT-105
+ 0.1 nM R1881
10 μM
μM - 0.3 1 3
- 1 10
- Enza Bical
AR
Actin
AR/Actin (Fold)
1 0.9 0.3 0.3
1 0.2 0.1
1 1.1 1.2

Figure 2. A. Intrinsic fluorescence assay using purified AR activation function-1 (AF-1) region of the NTD and DAARIs, UT-34 and UT-105. **B**. DAARIs degrade AR in PC preclinical models. LNCaP cells were treated with DAARIs, and Western for AR was performed.

Androgen-Glucuronidating Enzymes are Lower in Enzalutamide-Resistant Prostate Cancer Cells and Tumors

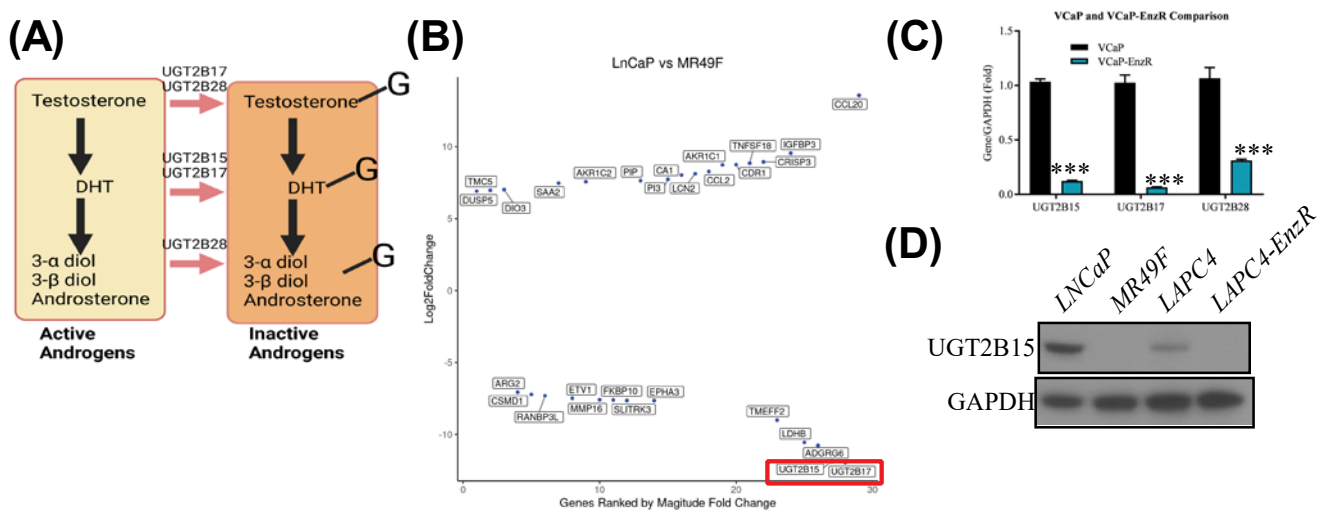
(A)
Testosterone
DHT
3-α diol
3-β diol
Androsterone
UGT2B17
UGT2B28
UGT2B15
UGT2B17
UGT2B28
G
Active Androgens
Inactive Androgens
(B)
LnCaP vs MR49F
Log2FoldChange
Genes Ranked by Magitude Fold Change
UGT2B15
UGT2B17
(C)
VCaP and VCaP-EnzR Comparison
VCaP
VCaP-EnzR
Gene/GAPDH (Fold)
UGT2B15
UGT2B17
UGT2B28
(D)
LNCaP
MR49F
LAPC4
LAPC4-EnzR
UGT2B15
GAPDH

Figure-1: **A**. Androgen metabolism pathway. **B**. Microarray analysis was performed with RNA extracted from LNCaP and MR49F (enza-resistant LNCaP) cells. Top 20 up- and down- regulated genes with UGT2B15 and UGT2B17 indicated by a box. **C.** mRNA expression of UGT2B in VCaP and enza-resistant VCaP. **D. UGT2B15 protein expression is lower in enza-resistant cells**. Western blot for UGT2B15 and GAPDH in parental and enza-resistant cells.

AR Antagonists and DAARIs Have Distinct Temporal Effects on AR Function. AR Antagonists, But Not DAARIs, Increase UGT2Bs.

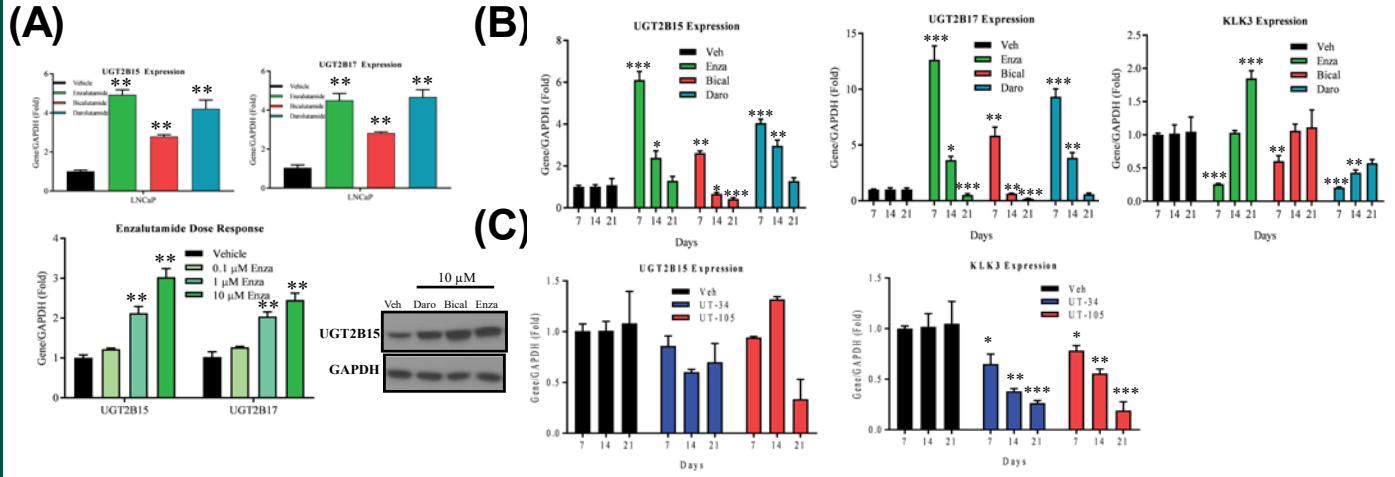
(A)
UGT2B15 Expression
UGT2B17 Expression
Enzalutamide Dose Response
Vehicle
0.1 μM Enza
1 μM Enza
10 μM Enza
10 μM
Veh Daro Bical Enza
UGT2B15
GAPDH
(B)
UGT2B15 Expression
UGT2B17 Expression
KLK3 Expression
Veh
Enza
Bical
Daro
Days
(C)
UGT2B15 Expression
KLK3 Expression
Veh
UT-34
UT-105
Days

Figure-3. AR antagonists increase UGT2Bs expression. **A**. LNCaP cells were grown in 3D cultures and were treated for four days. Expression of UGT2Bs was determined at the mRNA and protein levels. **B and C**. Experiments were performed in LNCaP cells as indicated in panel A with a time-course.

Results. LBD-binding AR inhibitors enzalutamide, darolutamide, and bicalutamide induce UGT2Bs at the mRNA and protein levels. The induction of UGT2Bs and inhibition of AR-target gene KLK3 were maximum between days 4 and 7 and no effect observed by day 21. Alternatively, NTD-binding DAARIs UT-34 and UT-105 elicited a sustained inhibition of KLK-3 despite their lack of effect on UGT2Bs expression.

Knock-Down of UGT2Bs Impairs AR Antagonist Efficacy

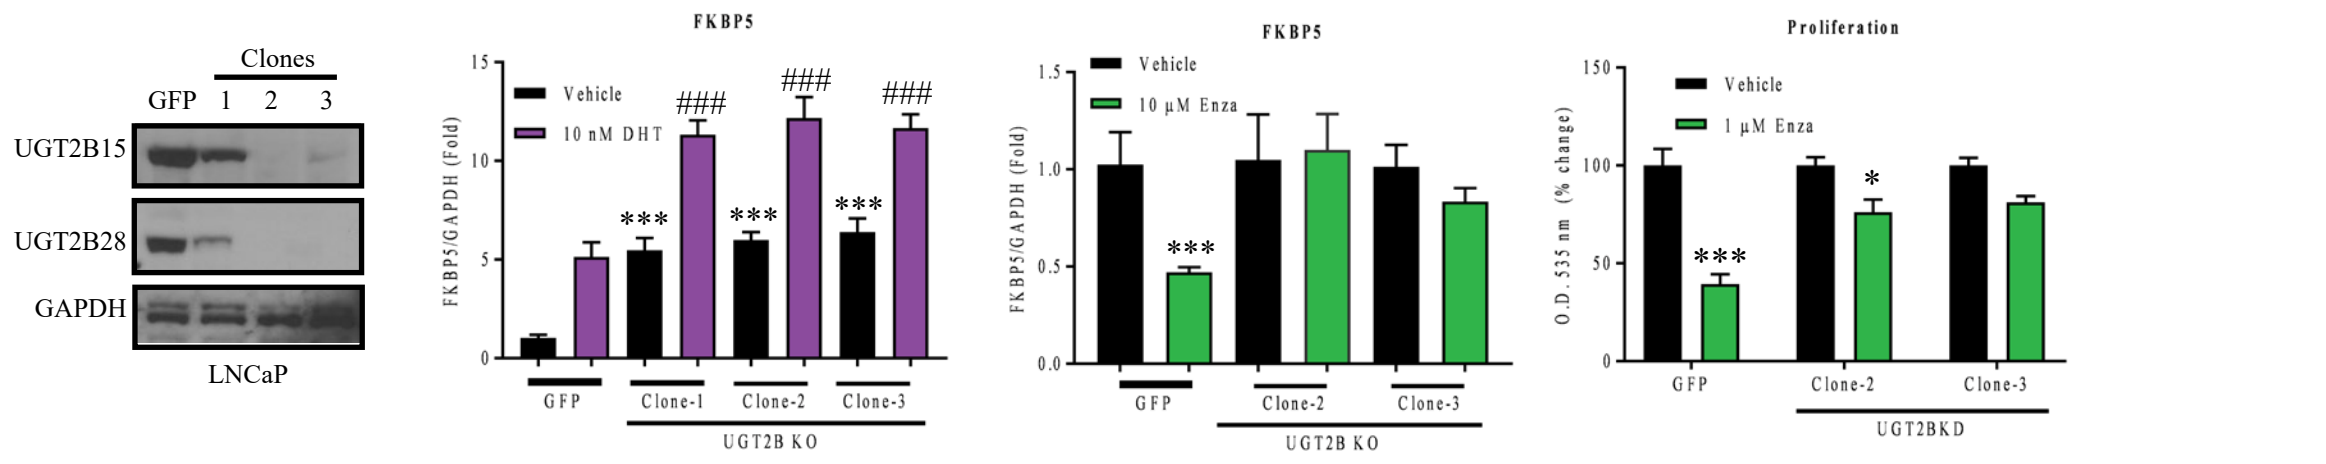
Clones
GFP 1 2 3
UGT2B15
UGT2B28
GAPDH
LNCaP
FKBP5
Vehicle
10 nM DHT
FKBP5/GAPDH (Fold)
GFP
Clone-1
Clone-2
Clone-3
UGT2B KO
FKBP5
Vehicle
10 μM Enza
GFP
Clone-2
Clone-3
UGT2B KO
Proliferation
Vehicle
1 μM Enza
O.D. 535 nm (% change)
GFP
Clone-2
Clone-3
UGT2BKD

Figure-4: Knock-down of UGT2Bs impairs canonical AR inhibitor efficacy. UGT2B15, 17, and 28 were knocked-down in LNCaP cells. Gene expression and cell proliferation in the presence of DHT or enzalutamide were measured in parental and knocked-down cells. GFP was used as a control.

AR Antagonists, But Not DAARIs, Efficacy In Vivo Correlates with UGT2B Expression

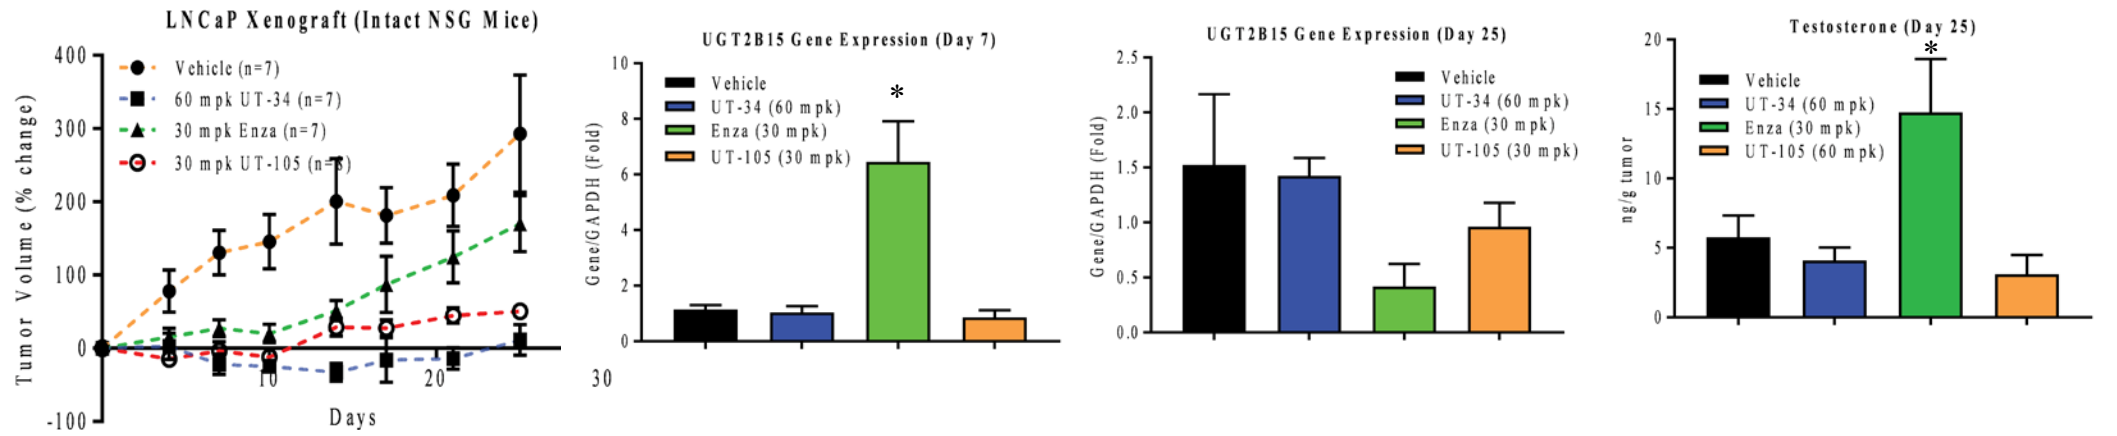
LNCaP Xenograft (Intact NSG Mice)
Vehicle (n=7)
60 mpk UT-34 (n=7)
30 mpk Enza (n=7)
30 mpk UT-105 (n=8)
Tumor Volume (% change)
Days
UGT2B15 Gene Expression (Day 7)
Vehicle
UT-34 (60 mpk)
Enza (30 mpk)
UT-105 (30 mpk)
Gene/GAPDH (Fold)
UGT2B15 Gene Expression (Day 25)
Vehicle
UT-34 (60 mpk)
Enza (30 mpk)
UT-105 (30 mpk)
Testosterone (Day 25)
Vehicle
UT-34 (60 mpk)
Enza (30 mpk)
UT-105 (60 mpk)
ng/g tumor

P value from vehicle (one-way ANOVA followed by TUKEY test)							
Days	4	7	10	14	17	21	25
UT-34	.001	.0062	.0001	.0032	.007	.002	.001
Enza		0.057	.011	.029			
UT-105	.023	.0042	.0005	.006	.042	.023	.0151

Figure-5: LNCaP xenograft was performed in NSG mice with enzalutamide and DAARIs. UGT2Bs expression was quantified on days 7 (in a subset of animals) and 25 (terminal sacrifice) of the treatment. Tumor testosterone was quantified by LC-MS/MS method.

Results: Enzalutamide was partially effective early in the treatment, which correlated with an increase in UGT2B15 expression. Long-term treatment with enzalutamide causes UGT2Bs to be inhibited, resulting in an increase in tumor androgen and lack of tumor growth inhibition. DAARIs provide a sustained tumor growth inhibition and do not depend on UGT2Bs.

Conclusion: Canonical LBD-targeting AR inhibitors induce expression of the androgen-metabolizing UGT2B enzymes, which might contribute to their anti-tumor activity. However, over time UGT2B expression levels are suppressed, leading to loss of canonical AR antagonist activity. NTD-targeting DAARIs are unaffected by UGT2B-mediated changes in androgen metabolism and retain in vivo efficacy independent of androgen metabolism.

Disclosure: SP, DH, YH, DDM, and RN are inventors in DAARI patents. RN is a consultant to Oncternal Therapeutics, Inc., San Diego. DAARI program is licensed to Oncternal Therapeutics, Inc. JBB and GFK are employees of Oncternal and receives pay and holds equity.

Funding: R01 CA229164 and Oncternal therapeutics Inc. San Diego, CA.